Demographic and Clinical Characteristics of Patients With SLE Across 5 Registries – the LupusNet Federated Data Network

Federico Zazzetti,* Urbano Sbarigia, William Giovanini, Claire Blacketer, Michel van Speybroeck, Teresa A Simon, Guo Li, Chetan S Karyekar, Reyhan Sonmez, Manuel F Ugarte-Gil, Rocio V Gamboa-Cardenas, Victor Pimentel-Quiroz, Guillermo Pons-Estel, Rosana Quintana, Verónica Saurit, Odirlei Andre Monticielo, Katiuzka Zuñiga, Jorge A Esquivel-Valerio, Martin Rebella, Cecilia Nora Pisoni, Francinne Machado Ribeiro, Carlos Núñez Álvarez, Kaleb Michaud, Patricia Katz, Rangi Kandane-Rathnayake, Eric Morand, Worawit Louthrenoo, Yi-Hsing Chen, Jiacai Cho, Laniyati Hamijoyo, Shue Fen Luo, Yeong-Jian Jan Wu, Sandra Navarra, Sargunan Sockalingam, Masayoshi Harigai, Zhuoli Zhang, Duminda Basnayake, Madelynn Chan, Tsutomu Takeuchi, Sang-Cheol Bae, Fiona Goldblatt, Sean O'Neill, Kristine (Pek Ling) Ng, Yih Jia Poh, Nicola Tugnet, Sunil Kumar, Michael Tee, Yoshiya Tanaka, Chak Sing Lau, Alberta Hoi, Mandana Nikpour, Mark Sapsford, Iñigo Rúa-Figueroa, José María Pego-Reigosa, María Galindo-Izquierdo, Jaime Calvo Alén, Antonio Fernández-Nebro, Raúl Menor Almagro, Frederic Lavie

*Presenting author.

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

0136

Key Takeaways

- LupusNet is currently the largest global SLE database and is the only database to connect registries across 4 continents, standardizing real-world data from existing registries and using a highly innovative, federated data network approach to allow better understanding of disease heterogeneity, patient populations, and treatment patterns, leading to improved outcomes for patients with SLE across the globe
- Across the LupusNet registries, there is variability in disease activity, as measured by PGA and SLEDAI, and in damage accrual, as measured by SDI
- The observed variability between registries may be related to differences in recruitment strategy, treatment strategy/access, health care systems, and patient race/ethnicity. Mean disease duration and treatments received prior to registration may also impact disease manifestations
- Results from this large, ongoing study will allow better understanding of disease heterogeneity, patient populations, and treatment patterns, with the goal of improving outcomes for patients with SLE across the globe

Background

Systemic lupus erythematosus (SLE) is a chronic, heterogeneous, autoantibody-driven disease associated with significant disease burden and high mortality risk[1,2]

– The epidemiology and clinical presentation of SLE differ across patient groups, with great variations in patient characteristics, such as age, sex, race, and ethnicity

Real-world evidence pertaining to SLE is dispersed among several global registries; therefore, no single registry collects all prevalent measures of SLE[3]

The Lupus Federated Data Network (LupusNet) is an interdisciplinary initiative that aims to combine and standardize data from current localized SLE registries to establish a comprehensive, global SLE database with augmented patient counts and more consistent data

– LupusNet currently includes 5 longitudinal observational registries of patients with SLE across North America, South America, Europe, and Asia-Pacific: Rheumatology Department of the Hospital Guillermo Almenara Irigoyen (ALMENARA), Asia Pacific Lupus Collaboration (APLC), National Databank for Rheumatic Diseases (FORWARD), Grupo Latino Americano de Estudio de Lupus (GLADEL), and RELESSER

Objective

To describe demographic and clinical characteristics of patients with SLE included in the 5 registries participating in LupusNet

Methods

LupusNet Study Design

- Data were collected from patients with SLE included in 5 disease registries participating in LupusNet at any point in time
- Collected data included baseline demographics, clinical characteristics, disease activity based on the Physician Global Assessment (PGA) and the Systemic Lupus Erythematosus Disease Activity Index (SLEDAI), and damage accrual based on the Systemic Lupus International Collaborating Clinics (SLICC) Damage Index (SDI) during a 90-day window before and after registration into the registry
 - Registry-specific variables included family history, socioeconomic factors, and procedure data
- Observation periods also varied by registry, with all patients having cohort entry dates and exit criteria of death or loss to follow-up. The frequency of patient visits by registry is shown in **Figure 1**

FIGURE 1: Visit frequency and data collection in LupusNet registries

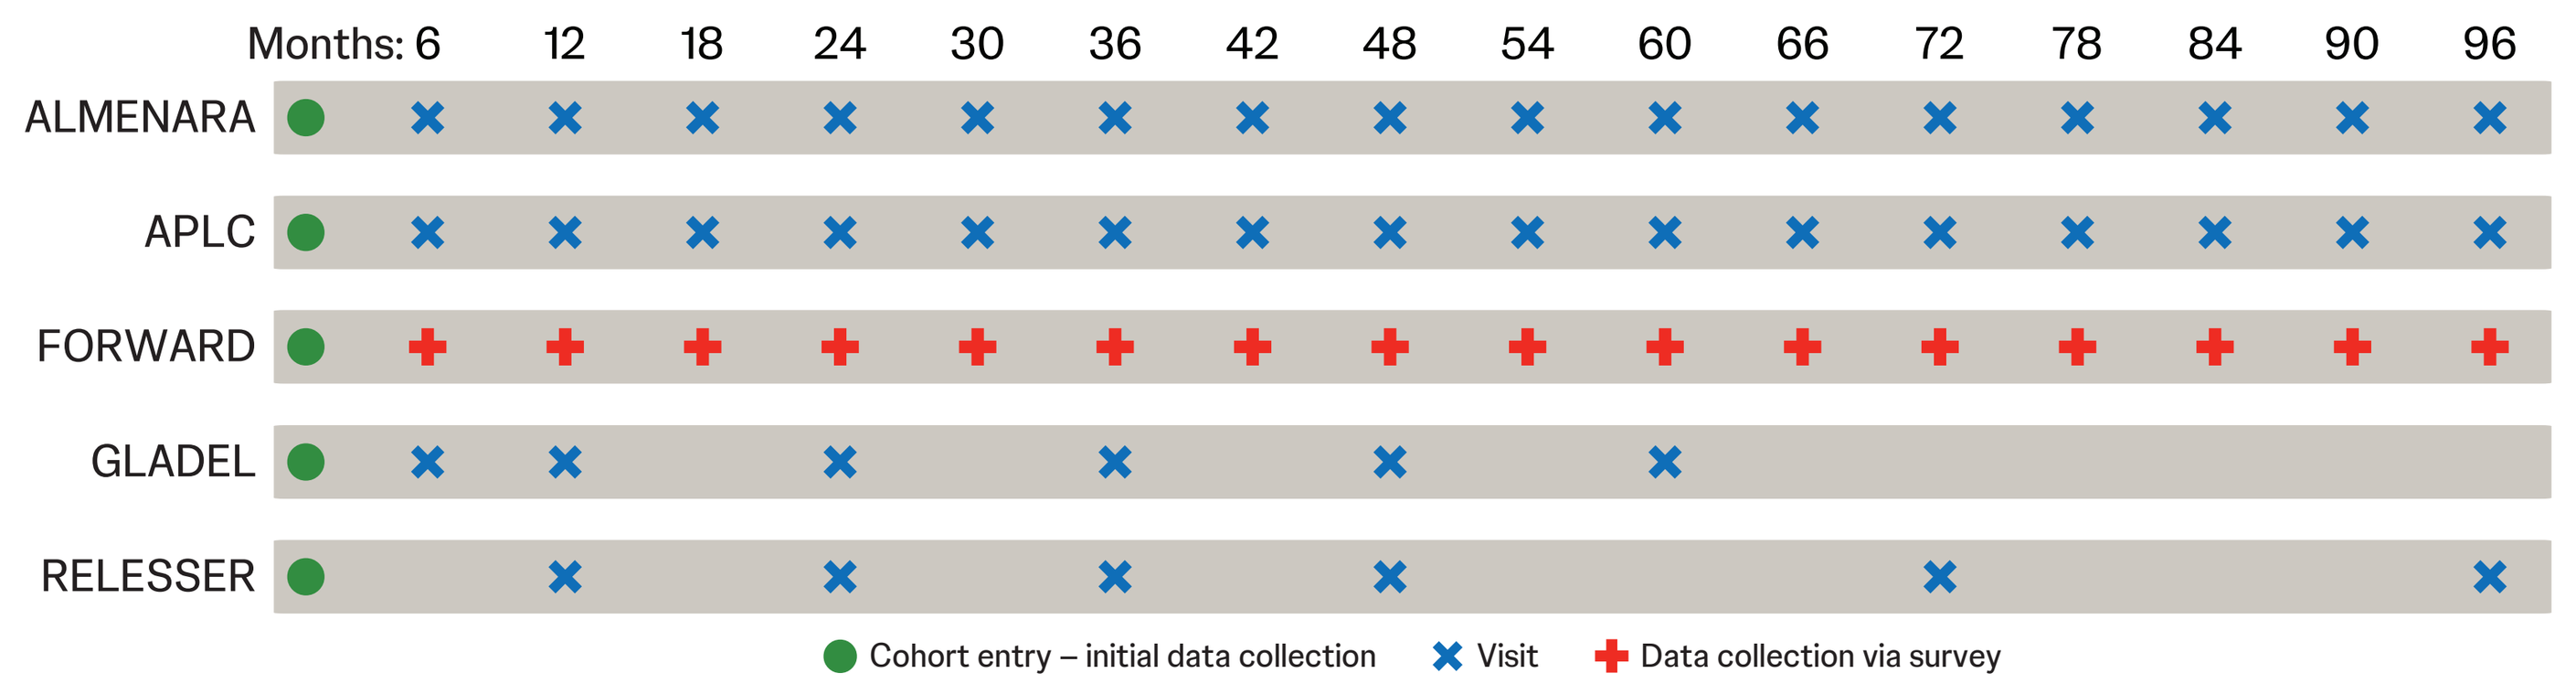
Months: 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96
ALMENARA
APLC
FORWARD
GLADEL
RELESSER
Cohort entry – initial data collection
Visit
Data collection via survey

ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus.

Study Analysis

- Registry datasets were harmonized using the Observational Medical Outcomes Partnership (OMOP) Common Data Model (CDM) v5.4
 - The OMOP CDM included a standard representation of health care experiences and common vocabularies for coding clinical concepts to enable consistent application of analyses across multiple data sources
 - The types of data eligible for standardization and harmonization in LupusNet vary depending on the registry design, which included differences in the timing of data collection, disease measurement or severity scales, and reporting of treatment duration; this was taken into consideration when analyzing results

Results

Characteristics of each registry participating in LupusNet are listed in Table 1

TABLE 1: Summary of registry populations

Registry	Regions/countries	Start year	Report type/data collection method	Inclusion criteria
ALMENARA	Peru	2012	Physician reported; data captured via standardized forms at visits occurring every 6 months	Patients aged ≥18 years who met ACR 1997 or SLICC 2012 criteria; patients with overlap syndrome (except APS and SjD) are excluded
APLC	Australia, China, Hong Kong, Indonesia, Japan, Korea, Malaysia, New Zealand, Philippines, Singapore, Sri Lanka, Taiwan, and Thailand	2013	Physician reported; data captured via standardized forms at visits occurring every 3-6 months. SDI and SF-36 are captured at annual visits only. Investigators at each site are responsible for data entry	Patients aged ≥18 years who met ACR 1997 or SLICC 2012 criteria
FORWARD	United States and Canada	1999	Patient reported; data captured via questionnaires completed by patients online or via paper forms every 6 months	Patients aged ≥18 years with a physician-confirmed diagnosis of SLE
GLADEL	South/Latin America (Argentina, Bolivia, Brazil, Chile, Colombia, Dominican Republic, Ecuador, Honduras, Mexico, Paraguay, Peru, Uruguay, and Venezuela)	2019	Physician reported; data captured via standardized forms at visits occurring annually for 7 years	Patients aged ≥18 years who met ACR 1982/1997 and/or SLICC 2012 criteria; patients with certain conditions were excluded[a] Four types of patients with SLE, based on the incidence and prevalence of lupus nephritis, were recruited
RELESSER	Spain	2007	Physician reported; data captured via standardized forms at visits occurring annually (first 5 visits), then biannually (final 2 visits)	Patients aged ≥16 years who met ACR 1997 or SLICC 2012 criteria or met 3 criteria with a diagnosis of SLE from an experienced rheumatologist The cohort includes patients with SLE within 0-18 months of diagnosis, with incomplete SLE, with serologically active clinically quiescent SLE, and with controls

[a]Patients were excluded from the GLADEL registry if they had any of the following: other systemic autoimmune diseases or overlap syndrome (rheumatoid arthritis, systemic sclerosis, dermatomyositis, systemic vasculitis, and others); urinary infection; pregnancy; or a history of hepatitis B, hepatitis C, or HIV infection. Patients presenting with APS associated with lupus were not excluded from the registry.
ACR=American College of Rheumatology, **ALMENARA**=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **APS**=antiphospholipid syndrome, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **HIV**=human immunodeficiency virus, **SDI**=SLICC Damage Index, **SF-36**=36-Item Short Form Survey, **SjD**=Sjögren's disease, **SLE**=systemic lupus erythematosus, **SLICC**=Systemic Lupus International Collaborating Clinics.

A total of 10,267 patients with SLE were included and mapped in LupusNet: 507 patients in South America, 3908 in Asia-Pacific, 3066 in North America, 980 in Central and South America, and 1806 in Europe. Demographic and clinical characteristics at registration are presented in Table 2

TABLE 2: Baseline demographic and clinical characteristics of patients in LupusNet[a,b]

Characteristic	ALMENARA (n = 507)	APLC (n = 3908)	FORWARD (n = 3066)	GLADEL (n = 980)	RELESSER (n = 1806)
Gender, female, n (%)	468 (92)	3597 (92)	2799 (91)	876 (89)	1625 (90)
Age at diagnosis, years, mean (SD)	35 (14)	31 (13)	36 (14)	29 (12)	31 (14)
Age at registry entry, years, mean (SD)	42 (13)	41 (13)	47 (14)	37 (12)	41 (14)
Duration from SLE diagnosis to registry entry, years, mean (SD)	7 (6)	10 (8)	11 (9)	8 (8)	10 (8)
Follow-up duration, years, mean (median)	6 (4)	3 (3)	4 (5)	1 (0.7)	4 (3)
Race, n (%)					
Asian	<10 (<2)	3442 (88)	–	–	–
Black or African American	<10 (<2)	–	345 (11)	78 (8)	–
White	<10 (<2)	–	1527 (50)	241 (25)	–
Other[c]	496 (98)	447 (11)	1194 (39)	654 (67)	–
Not available	–	–	–	–	1806 (100)
Ethnicity, n (%)					
Hispanic or Latino	–	<10 (<0.3)	144 (5)	–	–
Mixed ancestry	496 (98)	<10 (<0.3)	–	649 (66)	–
Other	11 (2)	3898 (100)	1051 (34)	331 (34)	–
Not Hispanic or Latino	–	–	1871 (61)	–	–
Not available	–	–	–	–	1806 (100)

[a]Absence of values for some categories is due to variations in data collection by the registries. [b]If <10 patients, the actual number is masked. [c]Includes patients with mixed ancestry and no matching concept, in addition to American Indian/Alaska Native patients in the FORWARD registry.
ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **SD**=standard derivation, **SLE**=systemic lupus erythematosus.

The mean (standard deviation [SD]) PGA score at registration ranged from 0.4 (0.5) in ALMENARA to 1.2 (1.0) in GLADEL from the 4 registries that collected these data. The majority of patients in the ALMENARA and APLC cohorts had mild or no disease activity, while more than a third of patients in GLADEL were classified as moderate or severe (Figure 2)

FIGURE 2: Disease severity based on PGA score at registration[a,b]

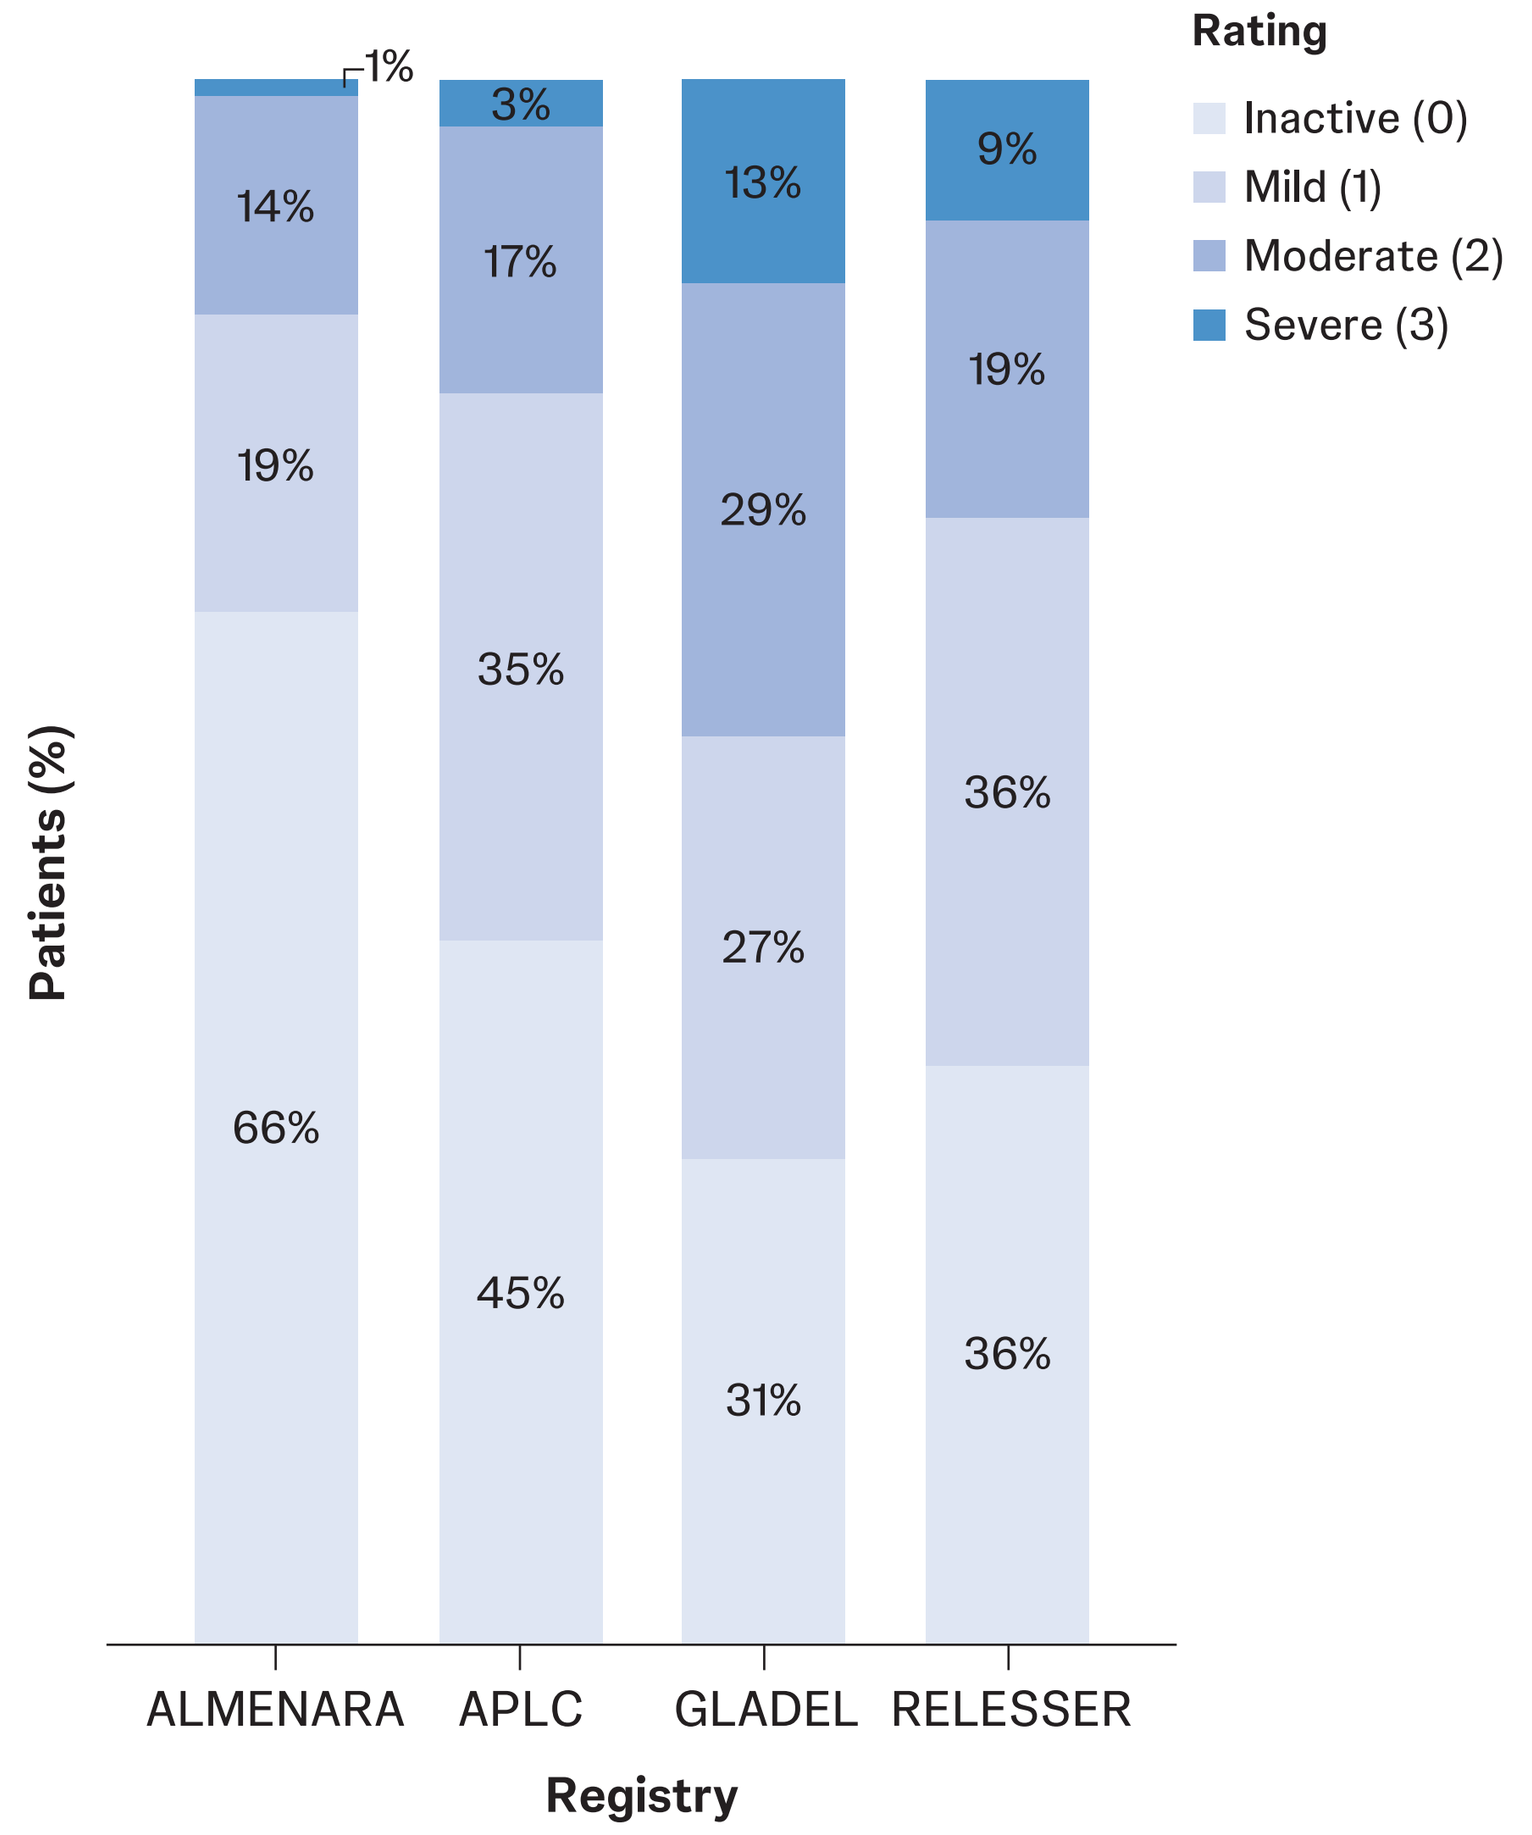
Rating
Inactive (0)
Mild (1)
Moderate (2)
Severe (3)
Patients (%)
1%
14%
19%
66%
3%
17%
35%
45%
13%
29%
27%
31%
9%
19%
36%
36%
ALMENARA
APLC
GLADEL
RELESSER
Registry

[a]The FORWARD registry does not collect these data. [b]341 patients from ALMENARA, 25 patients from APLC, and 787 patients from RELESSER did not have a PGA score recorded at registration. Percentages represent only patients with a PGA score recorded at registration.
ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **PGA**=Physician Global Assessment.

SLE disease activity by SLEDAI feature at registration was highly variable across registries, with the mean (SD) SLEDAI score ranging from 3.0 (4.1) to 7.2 (7.6; Figure 3)

FIGURE 3: SLEDAI-calculated total score distribution[a]

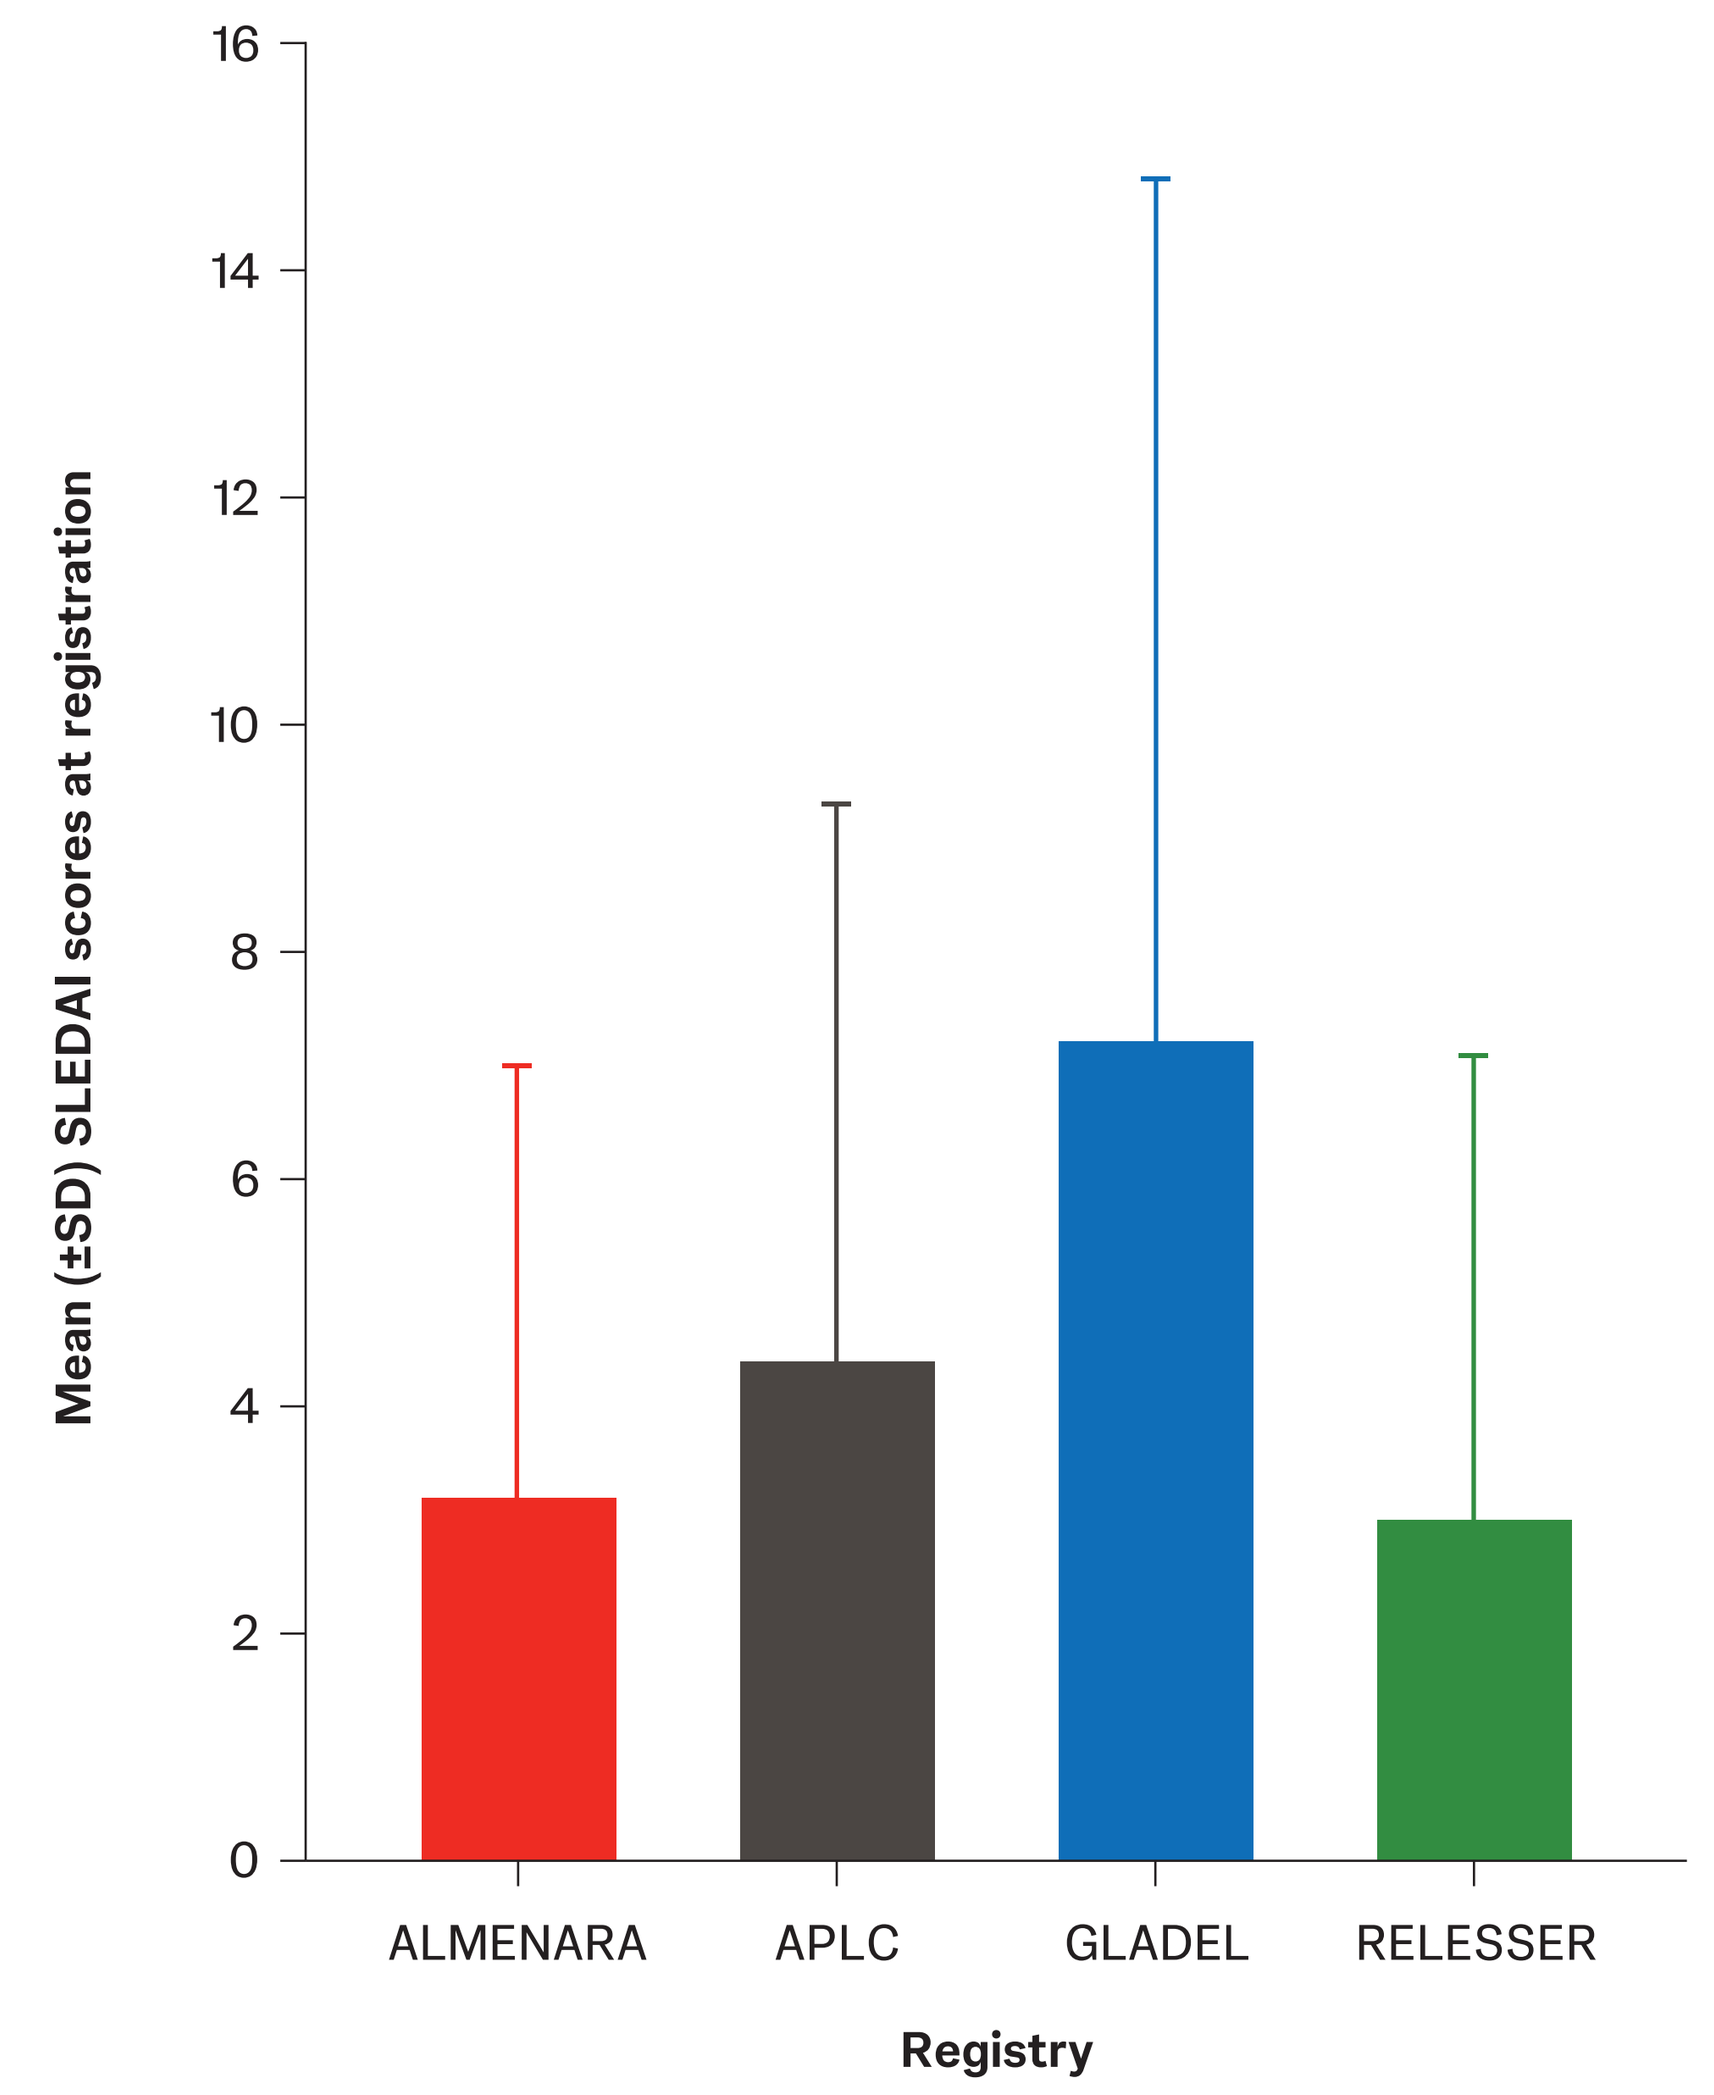
Mean (±SD) SLEDAI scores at registration
0
2
4
6
8
10
12
14
16
ALMENARA
APLC
GLADEL
RELESSER
Registry

[a]The FORWARD registry does not collect these data.
ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **SD**=standard deviation, **SLEDAI**=Systemic Lupus Erythematosus Disease Activity Index.

Heterogeneity in SLEDAI features was especially observed in arthritis (7%-21%), nephritis (1%-39%), DNA binding (7%-47%), and leukopenia (0%-14%; Figure 4)

FIGURE 4: Most frequent SLEDAI features at registration[a]

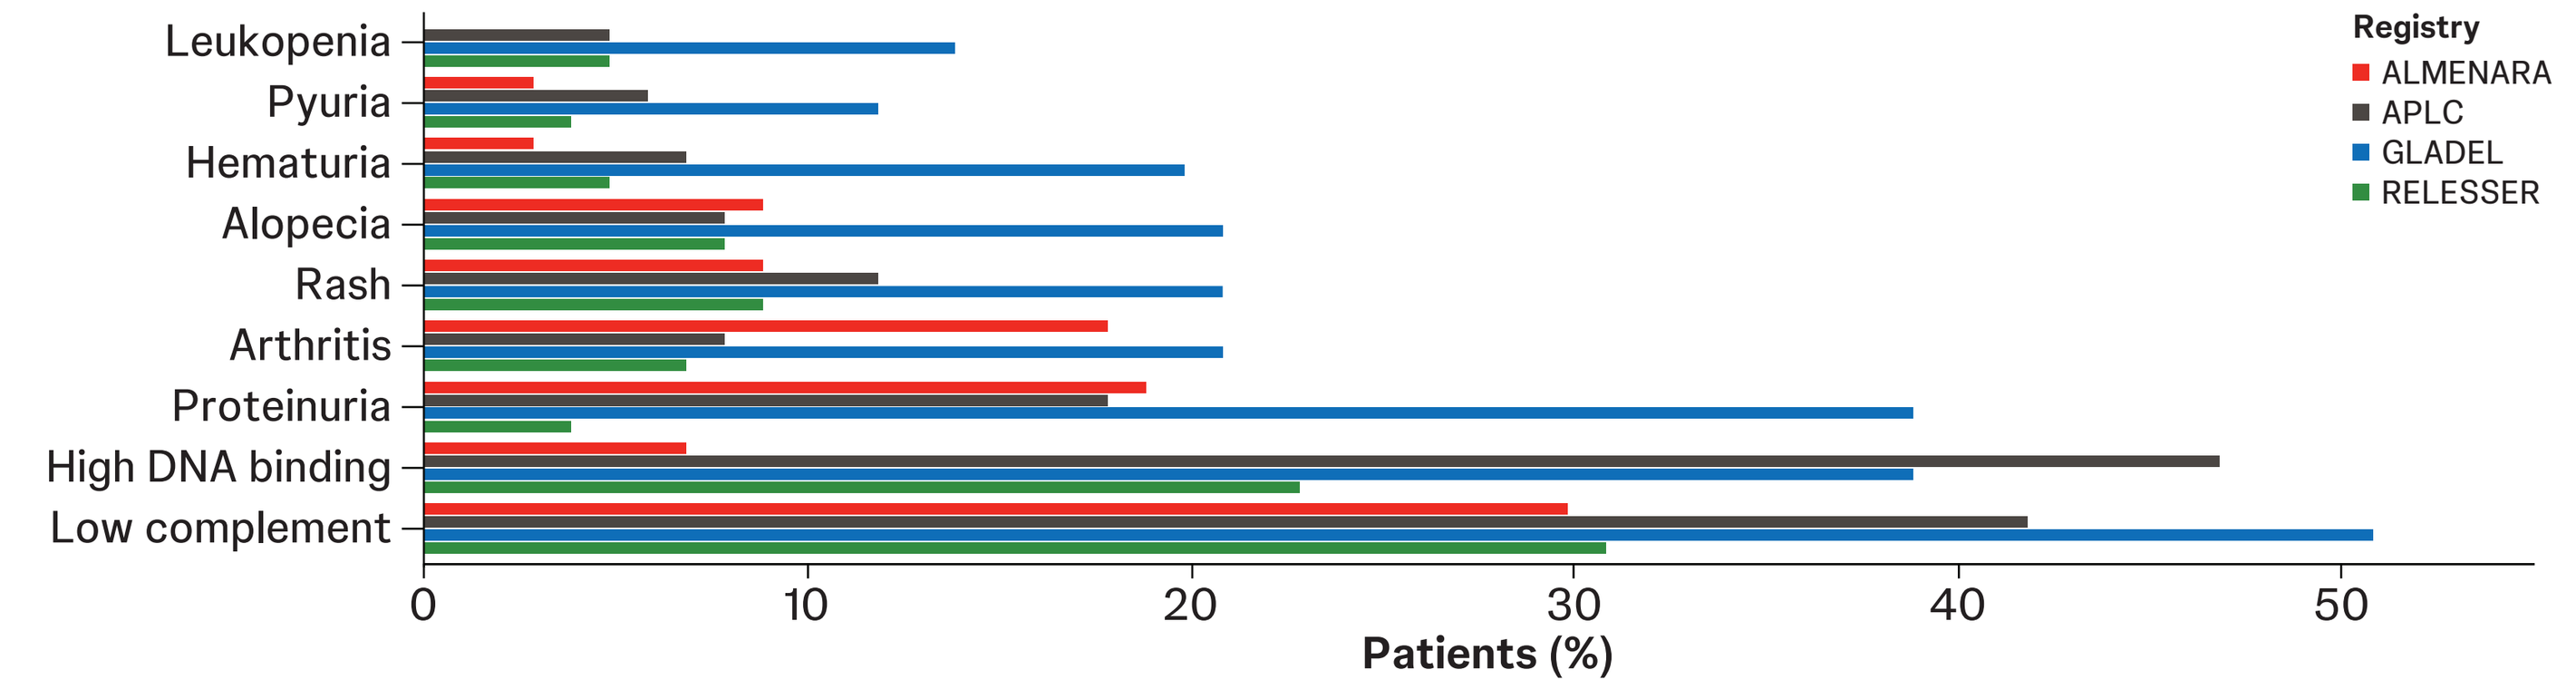
Leukopenia
Pyuria
Hematuria
Alopecia
Rash
Arthritis
Proteinuria
High DNA binding
Low complement
0
10
20
30
40
50
Patients (%)
Registry
ALMENARA
APLC
GLADEL
RELESSER

[a]The FORWARD registry does not collect these data.
ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **SLEDAI**=Systemic Lupus Erythematosus Disease Activity Index.

The mean (SD) SDI score at registration ranged from 0.6 (1.1) to 1.0 (1.4). The number and proportion of patients with each SDI characteristic are shown in Figure 5

- 11% of patients in ALMENARA had an estimated glomerular filtration rate <50%, 10% of patients in RELESSER had osteoporosis with fracture, and 27% of patients in GLADEL had proteinuria >3.5 g/24 hours due to specifically recruiting patients with lupus nephritis, leading to an overrepresentation of patients with renal features

FIGURE 5: Most common SDI characteristics at registration[a,b]

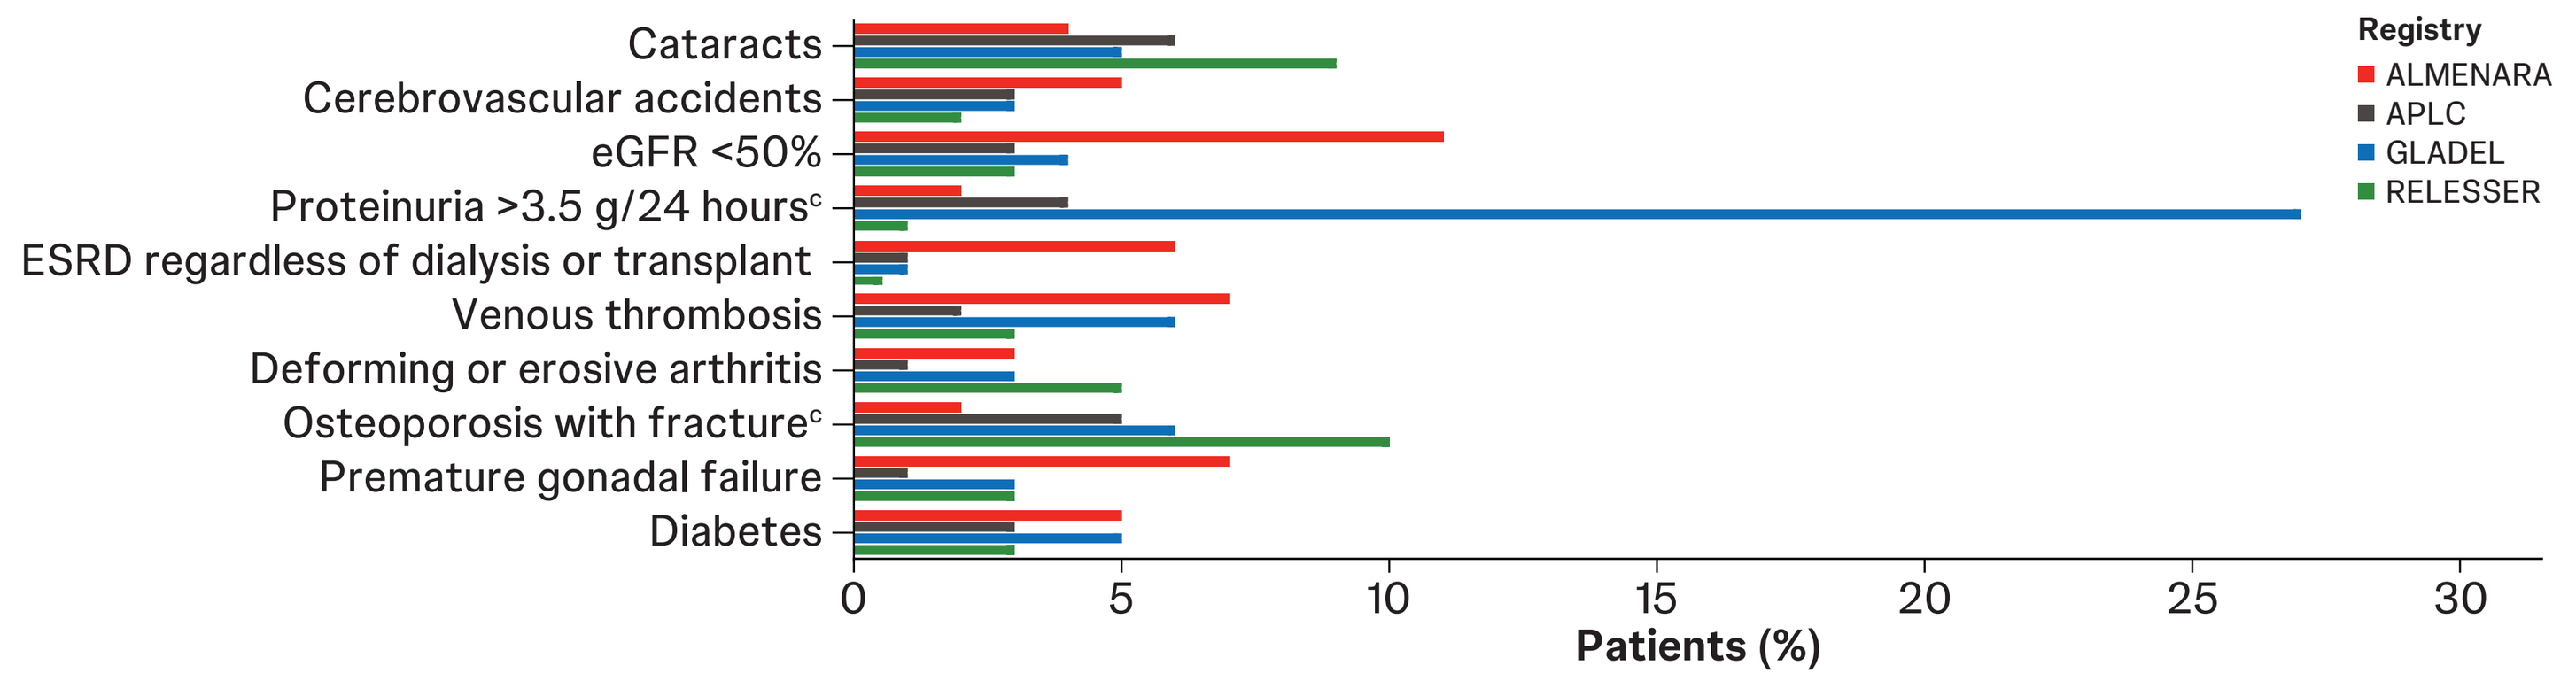
Cataracts
Cerebrovascular accidents
eGFR <50%
Proteinuria >3.5 g/24 hours[c]
ESRD regardless of dialysis or transplant
Venous thrombosis
Deforming or erosive arthritis
Osteoporosis with fracture[c]
Premature gonadal failure
Diabetes
0
5
10
15
20
25
30
Patients (%)
Registry
ALMENARA
APLC
GLADEL
RELESSER

[a]The FORWARD registry does not collect these data. [b]Features with an incidence <5% or with no incidence recorded across registries are not presented. [c]Incidence is <2% for ALMENARA.
ALMENARA=Rheumatology Department of the Hospital Guillermo Almenara Irigoyen, **APLC**=Asia Pacific Lupus Collaboration, **eGFR**=estimated glomerular filtration rate, **ESRD**=end-stage renal disease, **FORWARD**=National Databank for Rheumatic Diseases, **GLADEL**=Grupo Latino Americano de Estudio de Lupus, **SDI**=Systemic Lupus International Collaborating Clinics Damage Index.

PRESENTED AT: American College of Rheumatology (ACR) Convergence; November 14-19, 2024; Washington, DC, USA. **REFERENCES: 1.** Stojan G, Petri M. *Curr Opin Rheumatol.* 2018;30(2):144-150. **2.** Ameer MA, et al. *Cureus.* 2022;14(10):e30330. **3.** Lofland J, et al. *Arthritis Rheum.* 2019;71:2868-2869. Abstract 1611. **ACKNOWLEDGMENTS:** The authors wish to thank Bernardo Pons-Estel for his contributions to LupusNet. This study was sponsored by Janssen Research & Development, LLC. Medical writing support was provided by Aya Younes, PharmD, of Lumanity Communications Inc., and was funded by Janssen Global Services, LLC. **DISCLOSURES: FZ**, **US**, **WG**, **CB**, **MvS**, **TAS**, **GL**, **CSK**, **RS**, and **FL** are employees of Janssen and may hold stock/stock options in Johnson & Johnson. **MFU-G** received consulting and/or speaking fees from AstraZeneca, Ferrer, and GSK; and received research support from Janssen. **GP-E** received grants and consulting fees from and participated as a speaker, as an advisor, and/or on a steering committee for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics. **OAM** received speaking fees and/or honoraria from AbbVie, AstraZeneca, Celltrion, GSK, Janssen-Cliag, Novartis, and UCB. **RK-R** received research grants from GSK and Novartis. **SN** received consulting and speaking fees from Astellas, AstraZeneca, Aurinia, Biogen, and Idorsia. **TT** received grants from AbbVie, Chugai, and Mitsubishi-Tanabe; and received consulting and speaking fees and/or honoraria from AbbVie, Astellas, Chugai, Eli Lilly Japan, and Mitsubishi Tanabe. **KN** received speaking fees from Novartis. **YT** received speaking fees and/or honoraria from AbbVie, Asahi Kasei, Astellas, AstraZeneca, Boehringer Ingelheim, Chugai, Daiichi Sankyo, Eisai, Eli Lilly, Gilead, GSK, Pfizer, Taisho, and UCB; and received research grants from Boehringer Ingelheim, Chugai, and Taisho. **MS** received speaking fees from Novartis. **JMP-R** received speaker fees from and participated in advisory boards for AstraZeneca, GSK, and Otsuka; and received investigation grants from AstraZeneca and GSK. **AF-N** received consulting and speaking fees from AstraZeneca, Eli Lilly, Galapagos, Gebro Pharma, GSK, and Novartis; and received research grants from Argenx, AstraZeneca, Chemo, Galapagos, Janssen, Merck Serono, MSD, Novartis, Takeda, and UCB. **RVG-C**, **VP-Q**, **RQ**, **VS**, **KZ**, **JAE-V**, **MR**, **CNP**, **FMR**, **CNA**, **KM**, **PK**, **EM**, **WL**, **Y-HC**, **JC**, **LH**, **SFL**, **Y-JJW**, **SS**, **MH**, **ZZ**, **DB**, **MC**, **S-CB**, **FG**, **SO**, **YJP**, **NT**, **SK**, **MT**, **CSL**, **AH**, **MN**, **IR-F**, **MG-I**, **JCA**, and **RMA** declared no conflicts of interest.