ACR Convergence
Where Rheumatology Meets

POSTER ID
2417

GLADEL LATIN AMERICA
SCIENCE & EDUCATION

FREQUENCY AND ASSOCIATED FACTORS OF HERPES ZOSTER INFECTION IN SYSTEMIC LUPUS ERYTHEMATOSUS PATIENTS FROM LATIN-AMERICA

Romina Nieto, Lucia Hernández, Marina Scolnik, Subils Gisela Constanza, Verónica Saurit, Verónica Gabriela Savio, Valeria Arturi, Boris Kisluk, Luciana González Lucero, Wilfredo Patiño Grageda, María de los Ángeles Gargiulo, Odirlei Andre Monticielo, Ángela Luzia Branco Pinto Duarte, Eduardo F. Borba, Luciana Parente Costa Seguro, Edgard Torres dos Reis Neto, Oscar Neira, Gustavo Aroca Martinez, Antonio Iglesias Gamarra, Paul Méndez-Patarroyo, Rafael López Martínez, Margarita Portela Hernández, Carlos Núñez Álvarez, Yelitza C. González-Bello, Jorge Isaac Velasco Santos, Jorge A. Esquivel-Valerio, Marcos Aurelio Vázquez Báez, Maria Teresa Martinez de Filartiga, Magaly Alva Linares, Roberto Muñoz Louis, Carina Pizzarossa, Ana Carolina Ralle, María Camila Riascos, Joaquín Martínez Serventi, Graciela S. Alarcón, Bernardo Pons-Estel, Guillermo Pons-Estel on behalf of GLADEL.

BACKGROUND

Systemic lupus erythematosus (SLE) is an autoimmune disease with complex multi-systemic involvement. Herpes zoster (HZ) is caused by the reactivation of latent varicella-zoster virus (VZV) in patients who had been exposed earlier. HZ infection is most commonly seen in elderly and immunocompromised individuals, including those with autoimmune diseases such as rheumatoid arthritis and SLE. In Latin America, information about the estimated frequency and impact of HZ in patients with SLE is scarce. The aim of this study was to assess the epidemiology and clinical characteristics of HZ and to identify factors associated with the first HZ episode in SLE patients.

METHODS

GLADEL 2.0 is an observational multi-ethnic Latin-American SLE cohort. Forty-three centers from 10 Latin-American countries enrolled patients ≥18 years of age who fulfilled the 1982/1997 American College of Rheumatology (ACR) and/or the 2012 Systemic Lupus International Collaborating Clinics (SLICC) classification criteria. Baseline demographic, clinical, disease activity (SLEDAI), damage (SLICC/ACR Damage Index), laboratory and treatment data of patients with and without HZ events were examined. Continuous variables are presented as mean (±SD) or median (IQR) and categorical variables as count (percentage). Prevalence was calculated as the proportion of patients with HZ infection out of the SLE patients in the GLADEL 2.0 cohort. Associated factors were identified and logistic regression analysis was performed to examine the adjusted effects of these characteristics on the probability of experiencing at least one episode of HZ infection. The results are presented as ORs and their 95% CIs. P values < 0.05 were considered statistically significant. All analyses were performed with R v4.2.2 (or a later version).

RESULTS

Of the 1083 patients included in the GLADEL 2.0 cohort, 1073 were included in these analyses. A total of 83 HZ events were recorded at the baseline visit. The prevalence of HZ was 8.3% (CI: 6.8%-10.3%). SLE patients with history of HZ infection were more frequently female, with a higher frequency of cutaneous involvement (discoid lupus and alopecia), neurological involvement (psychosis and seizures), low complement, comorbidities and chronic renal failure (table 1). In terms of treatment, they also had a higher frequency of using methylprednisolone boluses, and immunosuppressants (IV cyclophosphamide, azathioprine, methotrexate, mycophenolate, rituximab and IV immunoglobulins). Multivariate analysis found that a history of psychosis and the use of methotrexate and mycophenolate were factors significantly associated with HZ events in these SLE patients (table 2).

Table 1: Sociodemographic, clinical and treatments according to Herpes Zoster events at cohort entry.

VARIABLE	HZ Ever (n=83)	HZ Never (n=990)	P VALUE[1]
Female, n %	79 (95.2)	883 (89.2)	0.092
Age at diagnosis (years), median (Q1, Q3)	25 (18-34.5)	27 (20-35)	0.097
Education level (years), median (Q1, Q3)	13 (12-16)	13 (11-16)	0.850
Ethnicity, n (%)			0.226
African Latin American	7 (8.4)	81 (8.2)	
Caucasian	26 (31.3)	248 (25.0)	
Mestizo	48 (57.8)	648 (65.5)	
Other	2 (2.4)	9 (0.9)	
Socioeconomic status, n(%)			0.305
Low/Medium low/Medium	58 (69.9)	749 (75.7)	
High/Medium high	25 (30.1)	225 (22.7)	
Medical insurance, n(%)	53 (63.9)	694 (70.1)	0.169
Cumulative clinical manifestations, n (%)			
Fever	39 (47.0)	401 (40.5)	0.296
Malar rash	59 (71.1)	595 (60.1)	0.060
Discoid lupus	13 (15.7)	74 (7.5)	0.019
Photosensitivity	57 (68.7)	606 (61.2)	0.239
Oral/nasopharyngeal ulcer	44 (53.0)	425 (42.9)	0.107
Alopecia	65 (78.3)	636 (64.4)	0.011
Arthritis	65 (78.3)	804 (81.2)	0.470
Pleuritis	25 (30.1)	253 (25.5)	0.365
Pericarditis	17 (20.5)	175 (17.7)	0.552
Persistent proteinuria	48 (57.8)	552 (55.8)	0.729
Cellular cylinders	25 (30.1)	245 (24.7)	0.175
Psychosis	7 (8.4)	29 (2.9)	0.017
Seizures	9 (10.8)	44 (4.4)	0.017
Hemolytic anemia	13 (15.7)	114 (11.5)	0.289
Leukopenia	41 (49.4)	449 (45.4)	0.647
Lymphopenia	53 (63.9)	521 (52.6)	0.085
Thrombocytopenia	25 (30.1)	219 (22.1)	0.135
Positive ANA	83 (100.0)	969 (97.9)	1.000
Anti-dsDNA positivity	67 (80.7)	742 (74.9)	0.583
Anti-Sm positivity	23 (27.7)	288 (29.0)	1.000
Lupus anticoagulant positivity	11 (13.3)	128 (12.9)	0.723
Anti-Cardiolipin positivity[a]	18 (21.7)	159 (16.1)	0.155
Anti-B2GPI positivity[b]	7 (8.4)	82 (8.3)	0.657
C3 low	71 (85.5)	730 (73.7)	0.043
C4 low	70 (84.3)	738 (74.5)	0.133
Comorbidities*	54 (65.1)	486 (49.1)	0.006
Chronic renal failure, n(%)	9 (10.8)	53 (5.4)	0.013
SLEDAI, median (Q1, Q3)	5 (2-10)	5 (1-12)	0.818
SDI, median (Q1, Q3)	0 (0-1)	0 (0-1)	0.055
Cumulative treatments, n(%)			
Antimalarials	83 (100)	950 (96.0)	0.111
Prednisone o equivalent	82 (98.8)	944 (95.4)	0.108
Methylprednisolone bolus	62 (74.7)	501 (50.6)	0.001
Immunosuppressants	78 (94.0)	817 (82.5)	0.005
IV Cyclophosphamide	42 (50.6)	340 (34.3)	0.010
Azathioprine	52 (60.6)	413 (41.7)	0.001
Methotrexate	29 (34.9)	222 (22.4)	0.026
Tacrolimus	5 (6.0)	36 (3.6)	0.163
Cyclosporin A	4 (4.8)	20 (2.0)	0.227
Mycophenolate mofetil	50 (60.2)	433 (43.7)	0.020
Rituximab	16 (19.3)	86 (8.7)	0.011
Belimumab	5 (6.0)	44 (4.4)	0.478
IV Immunoglobulin	8 (9.6)	29 (2.9)	0.006

[1]p-value corresponding to the Wilcoxon test for the comparison of 2 medians of quantitative variables or Fisher's Exact Test for qualitative variables as appropriate. *At least one of the following: diabetes mellitus, arterial hypertension, dyslipidemia. [a]IgA, IgG or IgM positivity with moderate or high titers (≥40 GPL or MPL) [b]IgA, IgG or IgM positivity. SLEDAI: Systemic Lupus Erythematosus Disease Activity Index; SDI: Systemic Lupus International Collaborating Clinics/American College of Rheumatology Disease Index

Table 2. Univariable and multivariable logistic regression analyses of factors associated with the occurrence of herpes zoster infection.

VARIABLE	Univariate Model Odds Ratio (OR)	P VALUE	Multivariate Model Odds Ratio (OR)	P VALUE
Gender, Female	0.42 (0.15, 1.16)	0.095	0.4 (0.11, 1.39)	0.148
Age at diagnosis	0.98 (0.96, 1.00)	0.083	1 (0.97, 1.02)	0.783
Ethnicity				
Caucasian	Ref		Ref	
African Latin American	0.82 (0.34, 1.97)	0.664	0.52 (0.18, 1.44)	0.207
Mestizo	0.71 (0.43, 1.16)	0.173	0.73 (0.41, 1.31)	0.294
Other	2.12 (0.43, 10.34)	0.353	2.93 (0.5, 17.2)	0.233
Malar rash	1.63 (1.00, 2.66)	0.052	1.28 (0.71, 2.31)	0.408
Discoid lupus	2.29 (1.21, 4.33)	0.011	1.6 (0.72, 3.58)	0.251
Alopecia	1.99 (1.16, 3.41)	0.012	1.52 (0.83, 2.81)	0.178
Persistent proteinuria	1.11 (0.70, 1.76)	0.647	0.84 (0.41, 1.7)	0.618
Psychosis	3.04 (1.29, 7.17)	0.011	2.91 (1.02, 8.27)	0.046
Seizures	2.61 (1.23, 5.55)	0.013	1.81 (0.69, 4.73)	0.225
Leukopenia	1.14 (0.73, 1.78)	0.574	0.87 (0.47, 1.6)	0.645
Lymphopenia	1.53 (0.96, 2.44)	0.073	1.45 (0.76, 2.76)	0.254
Low C3 or C4	1.85 (0.87, 3.91)	0.107	1.51 (0.64, 3.57)	0.351
Comorbidities*	1.92 (1.20, 3.06)	0.006	1.72 (0.99, 3)	0.055
Chronic renal failure	2.13 (1.01, 4.50)	0.046	2.05 (0.85, 4.96)	0.111
SDI at cohort entry	0.99 (0.96, 1.02)	0.526	0.98 (0.94, 1.02)	0.254
SLEDAI at cohort entry	1.51 (0.96, 2.38)	0.074	0.74 (0.42, 1.3)	0.291
Prednisone	3.30 (0.45, 24.35)	0.242	0.99 (0.12, 7.94)	0.993
IV Cyclophosphamide	1.92 (1.22, 3.01)	0.005	1.42 (0.74, 2.73)	0.292
Methotrexate	1.83 (1.14, 2.95)	0.012	1.89 (1.06, 3.38)	0.031
Azathioprine	2.32 (1.46, 3.68)	<0.001	1.51 (0.88, 2.59)	0.132
Mycophenolate mofetil	2.49 (1.55, 4.00)	<0.001	2.06 (1.1, 3.84)	0.023
Rituximab	2.49 (1.38, 4.48)	0.002	1.43 (0.68, 3.04)	0.347
IV Immunoglobulin	3.51 (1.55, 7.95)	0.003	2.5 (0.89, 7.05)	0.083

*At least one of the following: diabetes mellitus, arterial hypertension, dyslipidemia. SLEDAI: Systemic Lupus Erythematosus Disease Activity Index; SDI: Systemic Lupus International Collaborating Clinics/American College of Rheumatology Disease Index

CONCLUSIONS

In SLE patients from the GLADEL 2.0 cohort, the **prevalence** of HZ infection was found to be **less than 10%.**

Neurological compromise and the use of immunosuppressants such as **methotrexate** and **mycophenolate** were associated with the occurrence of these events.

It is important to be aware of the **risk of HZ in SLE patients.**

Future research may be able to establish predictive factors of HZ occurrence in these patients.

DISCLOSURES

GP-E received grants and consulting fees from and participated as a speaker and advisor to Abbvie, AstraZeneca, Boehringer-Ingelheim, GLADEL, GSK, Janssen, Novartis, Pfizer and Remegen. OAM received grants and consulting fees from and participated as a speaker and advisor to AbbVie/Abbott, Aspen, AstraZeneca, Bristol-Myers Squibb (BMS), GlaxoSmithKlein(GSK), Janssen, Novartis, Roche and UCB. ERN received grants and consulting fees from and participated as a speaker and advisor to AbbVie/Abbott, AstraZeneca, Bristol-Myers Squibb(BMS), GlaxoSmithKlein(GSK) and Novartis. RN, LH, MS, GCS, VS, VGS, VA, BK, LGL, WPG, MAG, ALBPD, EB, LS, ON, GAM, AIG, PMP, RL, MPD, CNA, YCG-B, JIVS, JAE-V, MV, MTMDF, MAL, RML, CP, ACR, MCR, JMS, GA and BP-E declared no conflicts of interest.

ACKNOWLEDGMENTS

This study was sponsored by GlaxoSmithKline Supported Studies Programme.